A pharmacoscintigraphic study of the relationship between tablet erosion and pharmacokinetics of oral semaglutide

1. **Alyson Connor**
Quotient Clinical, Nottingham, UK
2. **Morten Donsmark**
Novo Nordisk A/S, Søborg, Denmark
3. **Marie-Louise Hartoft-Nielsen**
Novo Nordisk A/S, Søborg, Denmark
4. **Flemming L Søndergaard**
Novo Nordisk A/S, Søborg, Denmark
5. **Tine A Bækdal**
Novo Nordisk A/S, Søborg, Denmark

Introduction

- When administered orally, peptide-based drugs are susceptible to degradation in the stomach due to exposure to low pH and proteolytic enzymes. Furthermore, absorption is compromised by the limited permeability of the gastrointestinal epithelium.
- Semaglutide, a glucagon-like peptide-1 (GLP-1) analog, has been co-formulated in a tablet with the absorption enhancer sodium N-(8-[2-hydroxybenzoyl] amino) caprylate (SNAC), in order to promote the absorption after oral administration.
- SNAC protects against proteolytic degradation of semaglutide through a localized increase in pH and facilitates the absorption of semaglutide across the gastrointestinal epithelium primarily via the transcellular route.

Objective

- To investigate the anatomical site of tablet erosion, the rate of tablet erosion and the gastrointestinal transit of oral semaglutide, and to correlate these parameters to the pharmacokinetic properties of oral semaglutide in order to elucidate if tablet erosion kinetics may influence the extent of absorption of oral semaglutide.
- To evaluate the effect of water volume administered with dosing on tablet erosion kinetics and pharmacokinetic properties of oral semaglutide.

Methods

Design

- This was a randomized, open-label, two-period, crossover trial conducted at Quotient Clinical, Nottingham, UK.
- Eligible subjects were healthy males, aged 18–64 years with a body mass index of 18.5–30.0 kg/m^2.
- Subjects received single doses of oral semaglutide (10 mg with 300 mg SNAC) administered with 50 mL and 240 mL of water, respectively, in two different treatment periods in randomized sequence (Figure 1). Dosing occurred in the fasted state and was followed by 4 hours post-dose fasting.

Figure 1 Trial design.

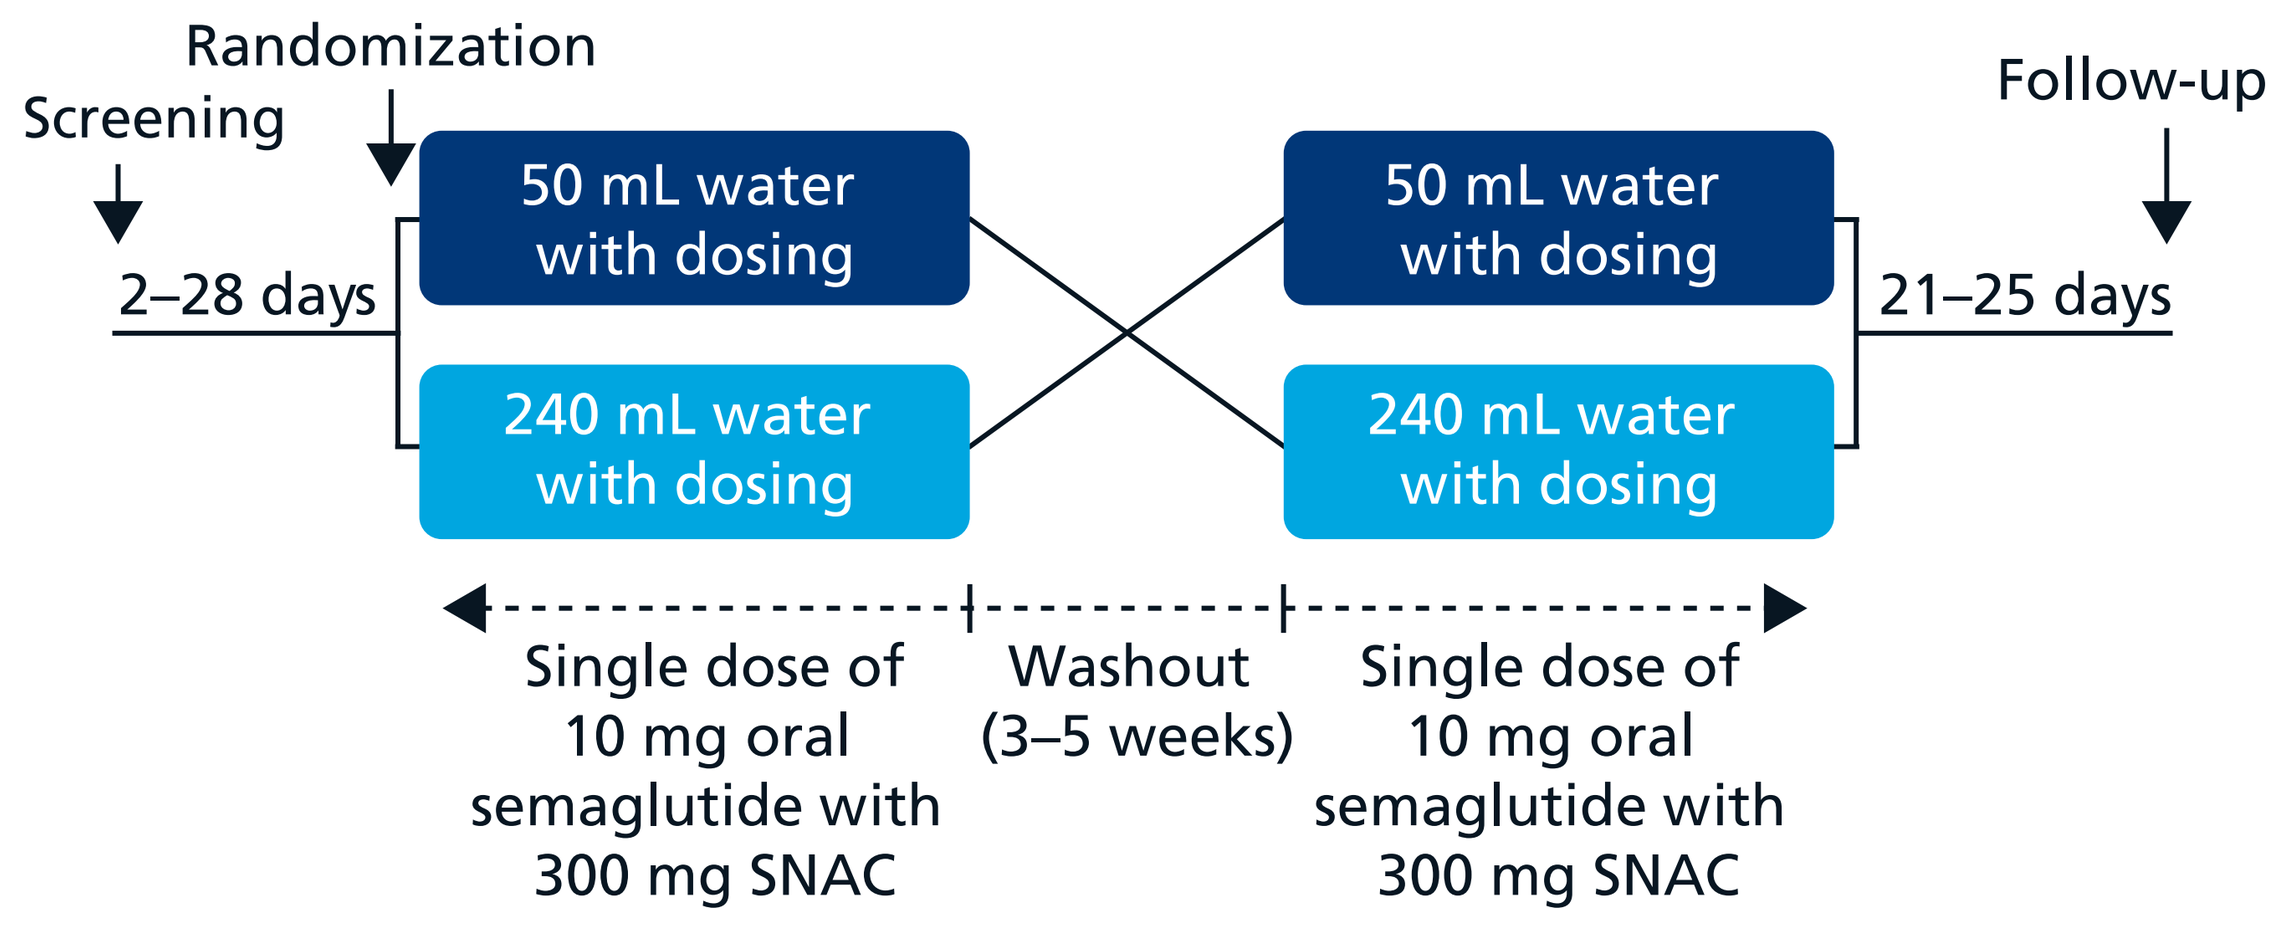

Randomization
Screening
2–28 days
50 mL water with dosing
240 mL water with dosing
50 mL water with dosing
240 mL water with dosing
Follow-up
21–25 days
Single dose of 10 mg oral semaglutide with 300 mg SNAC
Washout (3–5 weeks)
Single dose of 10 mg oral semaglutide with 300 mg SNAC

Scan here to access more Novo Nordisk scientific materials presented at ADA 2017

Figure 2 Gamma scintigraphic imaging of tablet erosion in the stomach from 15 to 120 minutes after a single dose of 10 mg oral semaglutide containing ^{111}In labelled ion-exchange resin in a representative healthy male subject.

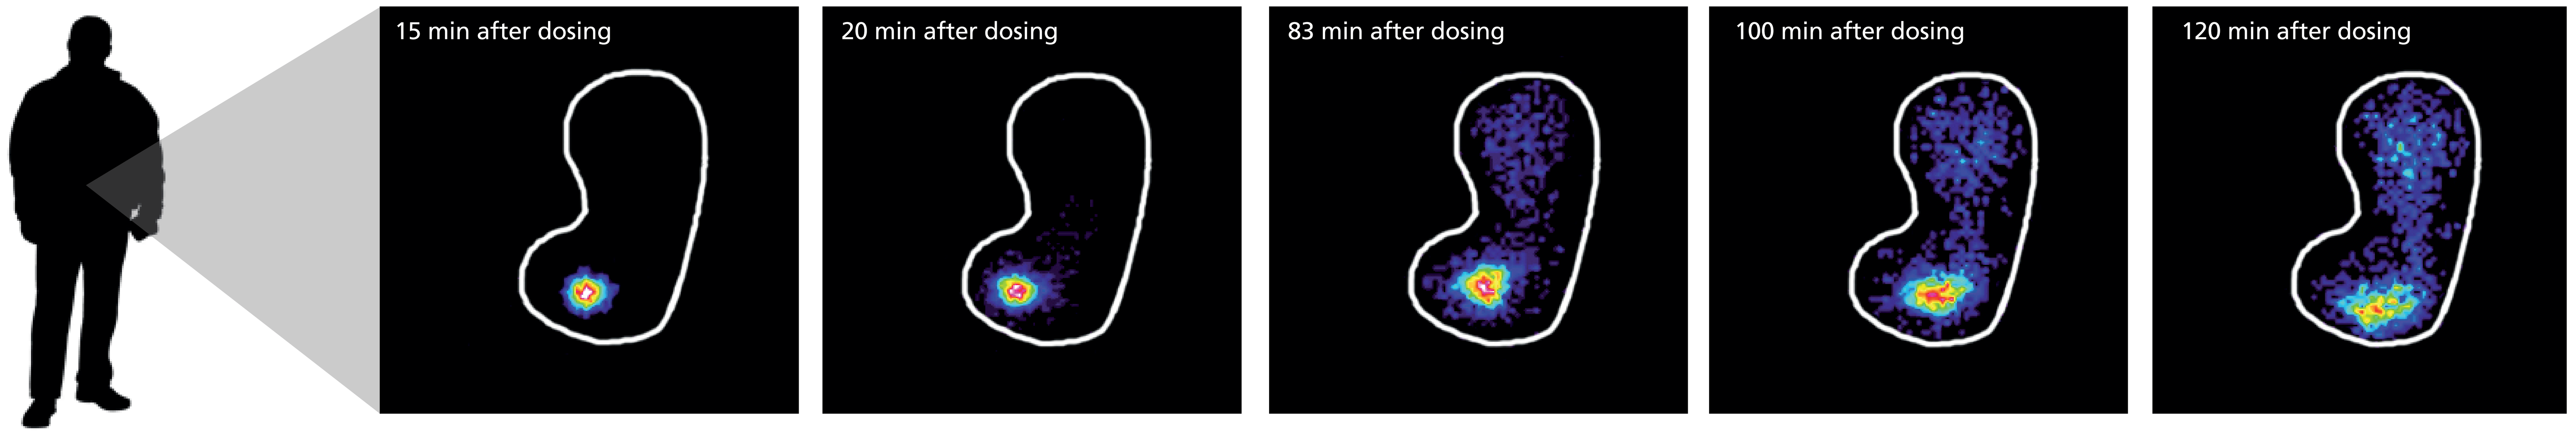

15 min after dosing
20 min after dosing
83 min after dosing
100 min after dosing
120 min after dosing

The intense colors within the stomach (e.g. red/yellow/green/blue) represent the tablet core and released radioactivity.

- Anatomical location of the tablet at the time of tablet erosion as well as gastrointestinal transit were assessed by gamma scintigraphy using a gamma camera (General Electric Maxicamera) with a 40 cm field of view.
- Oral semaglutide tablets contained ^{111}In labelled ion-exchange resin, and the water used for tablet administration was labelled with ^{99m}Tc (to provide an outline of the stomach).
- Dynamic imaging was performed during the first minute after dosing, while subjects were sitting. Subsequently, static images were recorded frequently until 4 hours after dosing, while subjects were standing (and allowed to sit or remain moderately active in between static imaging time points).
- Semaglutide concentrations were measured frequently in plasma until 24 hours after administration by means of a validated assay using plasma protein precipitation followed by liquid chromatography with tandem mass spectrometry detection.
- Safety assessments included adverse events, hypoglycemic episodes, physical examination, vital signs, electrocardiogram and laboratory safety variables.

Statistical analysis

- Time to complete tablet erosion (CTE) as well as plasma exposure ($AUC_{0–24h}$) and maximum concentration (C_{max}) of semaglutide were log-transformed and compared between the two water volumes using linear mixed models with water volume and period as fixed effects and subject as a random effect. The models for $AUC_{0–24h}$ and C_{max} allowed for left-censoring.
- To evaluate if time to CTE and time to complete gastric emptying of the tablet were correlated with semaglutide exposure, $AUC_{0–24h}$ and C_{max} were log-transformed and analyzed in linear mixed models with the log-transformed time to CTE or time to complete gastric emptying of the tablet as covariate, water volume and period as fixed effects and subject as a random effect.

Results

Subject characteristics

- A total of 26 healthy male subjects were included with a mean±standard deviation age of 38±11 years, body weight of 83.4±11.0 kg and body mass index of 25.9±2.3 kg/m^2.

Pharmacoscintigraphy

- CTE occurred in the stomach irrespective of water volume administered with the tablet. Representative scintigraphic images are presented in Figure 2.
- Time to CTE appeared to be approximately 50% longer after dosing with 50 mL vs. 240 mL water, although this was not statistically significant (estimated mean time to CTE 85 vs. 57 minutes; ratio 50/240 mL [95% confidence interval] 1.51 [0.96;2.37], p=0.072).
- Semaglutide plasma concentrations showed that systemic absorption of oral semaglutide occurred early independent of water volume with dosing (Figure 3). Median time to maximum semaglutide plasma concentration (t_{max}) was 1.5 hours with both water volumes.
- Semaglutide $AUC_{0–24h}$ and C_{max} were approximately 70% higher when dosed with 50 mL vs. 240 mL water (Figure 4).
- Higher semaglutide exposure (both $AUC_{0–24h}$ and C_{max}) was significantly correlated with slower tablet erosion and slower gastric emptying (all p<0.001).
- These results suggest that the higher semaglutide exposure levels observed with the 50 mL vs. the 240 mL water volume may be mediated through slower rate of tablet erosion and slower gastric emptying.

Figure 3 Estimated mean semaglutide plasma concentration-time profiles after a single dose of 10 mg oral semaglutide with 50 mL or 240 mL water in healthy male subjects.

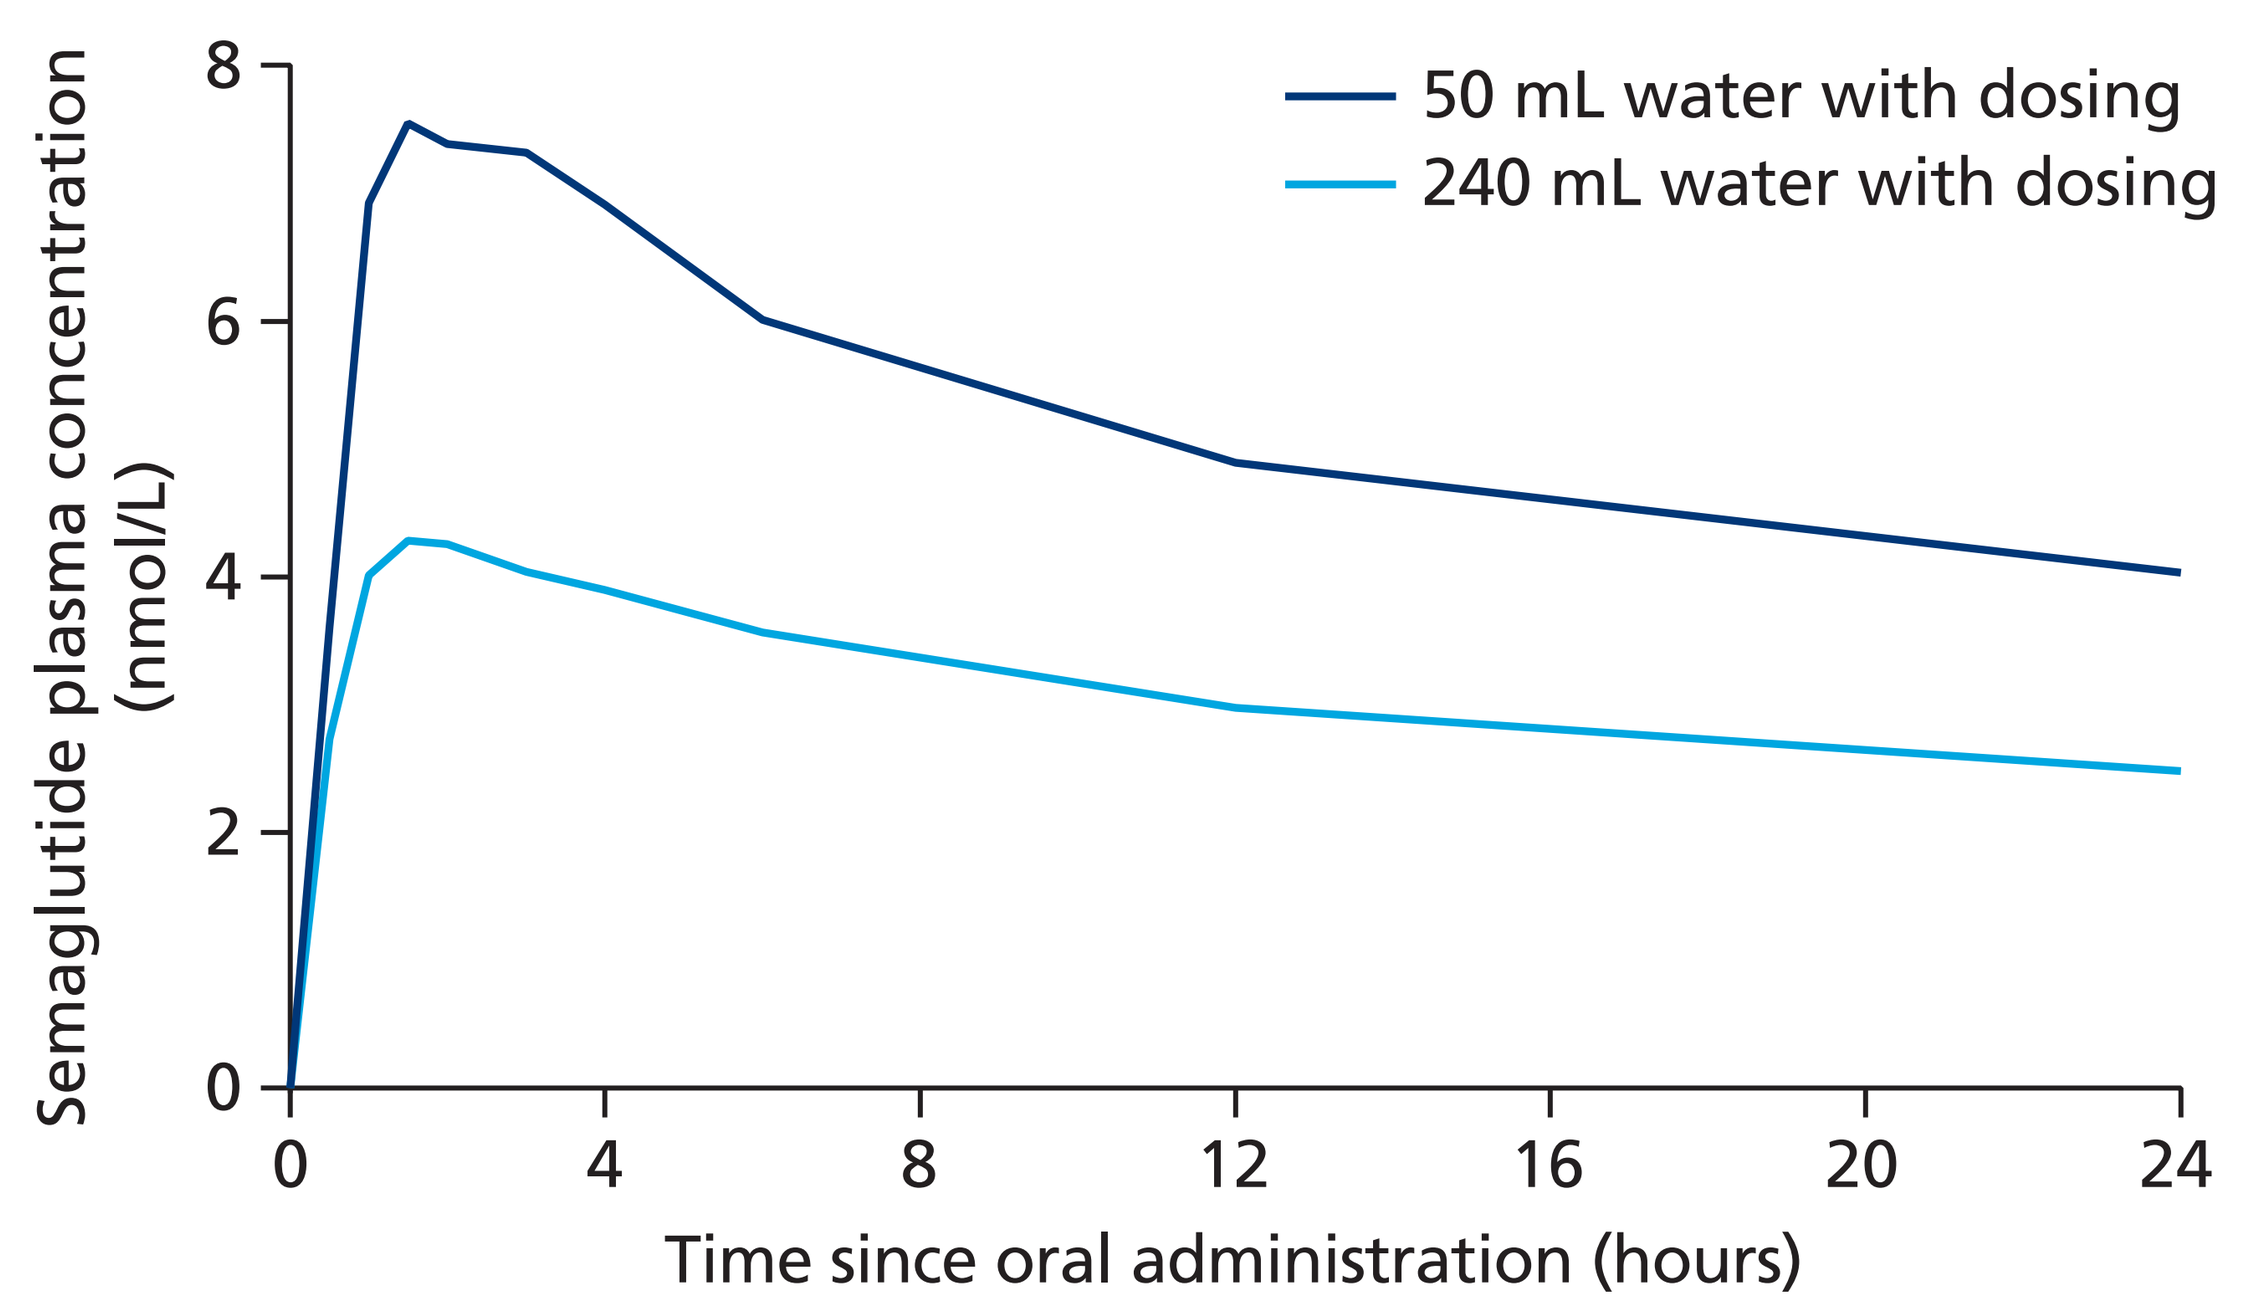

Semaglutide plasma concentration (nmol/L)
8
6
4
2
0
0
4
8
12
16
20
24
Time since oral administration (hours)
50 mL water with dosing
240 mL water with dosing

Figure 4 Effect of water volume with dosing on (A) AUC_{0-24h} and (B) C_{max} of semaglutide after a single dose of 10 mg oral semaglutide in healthy male subjects.

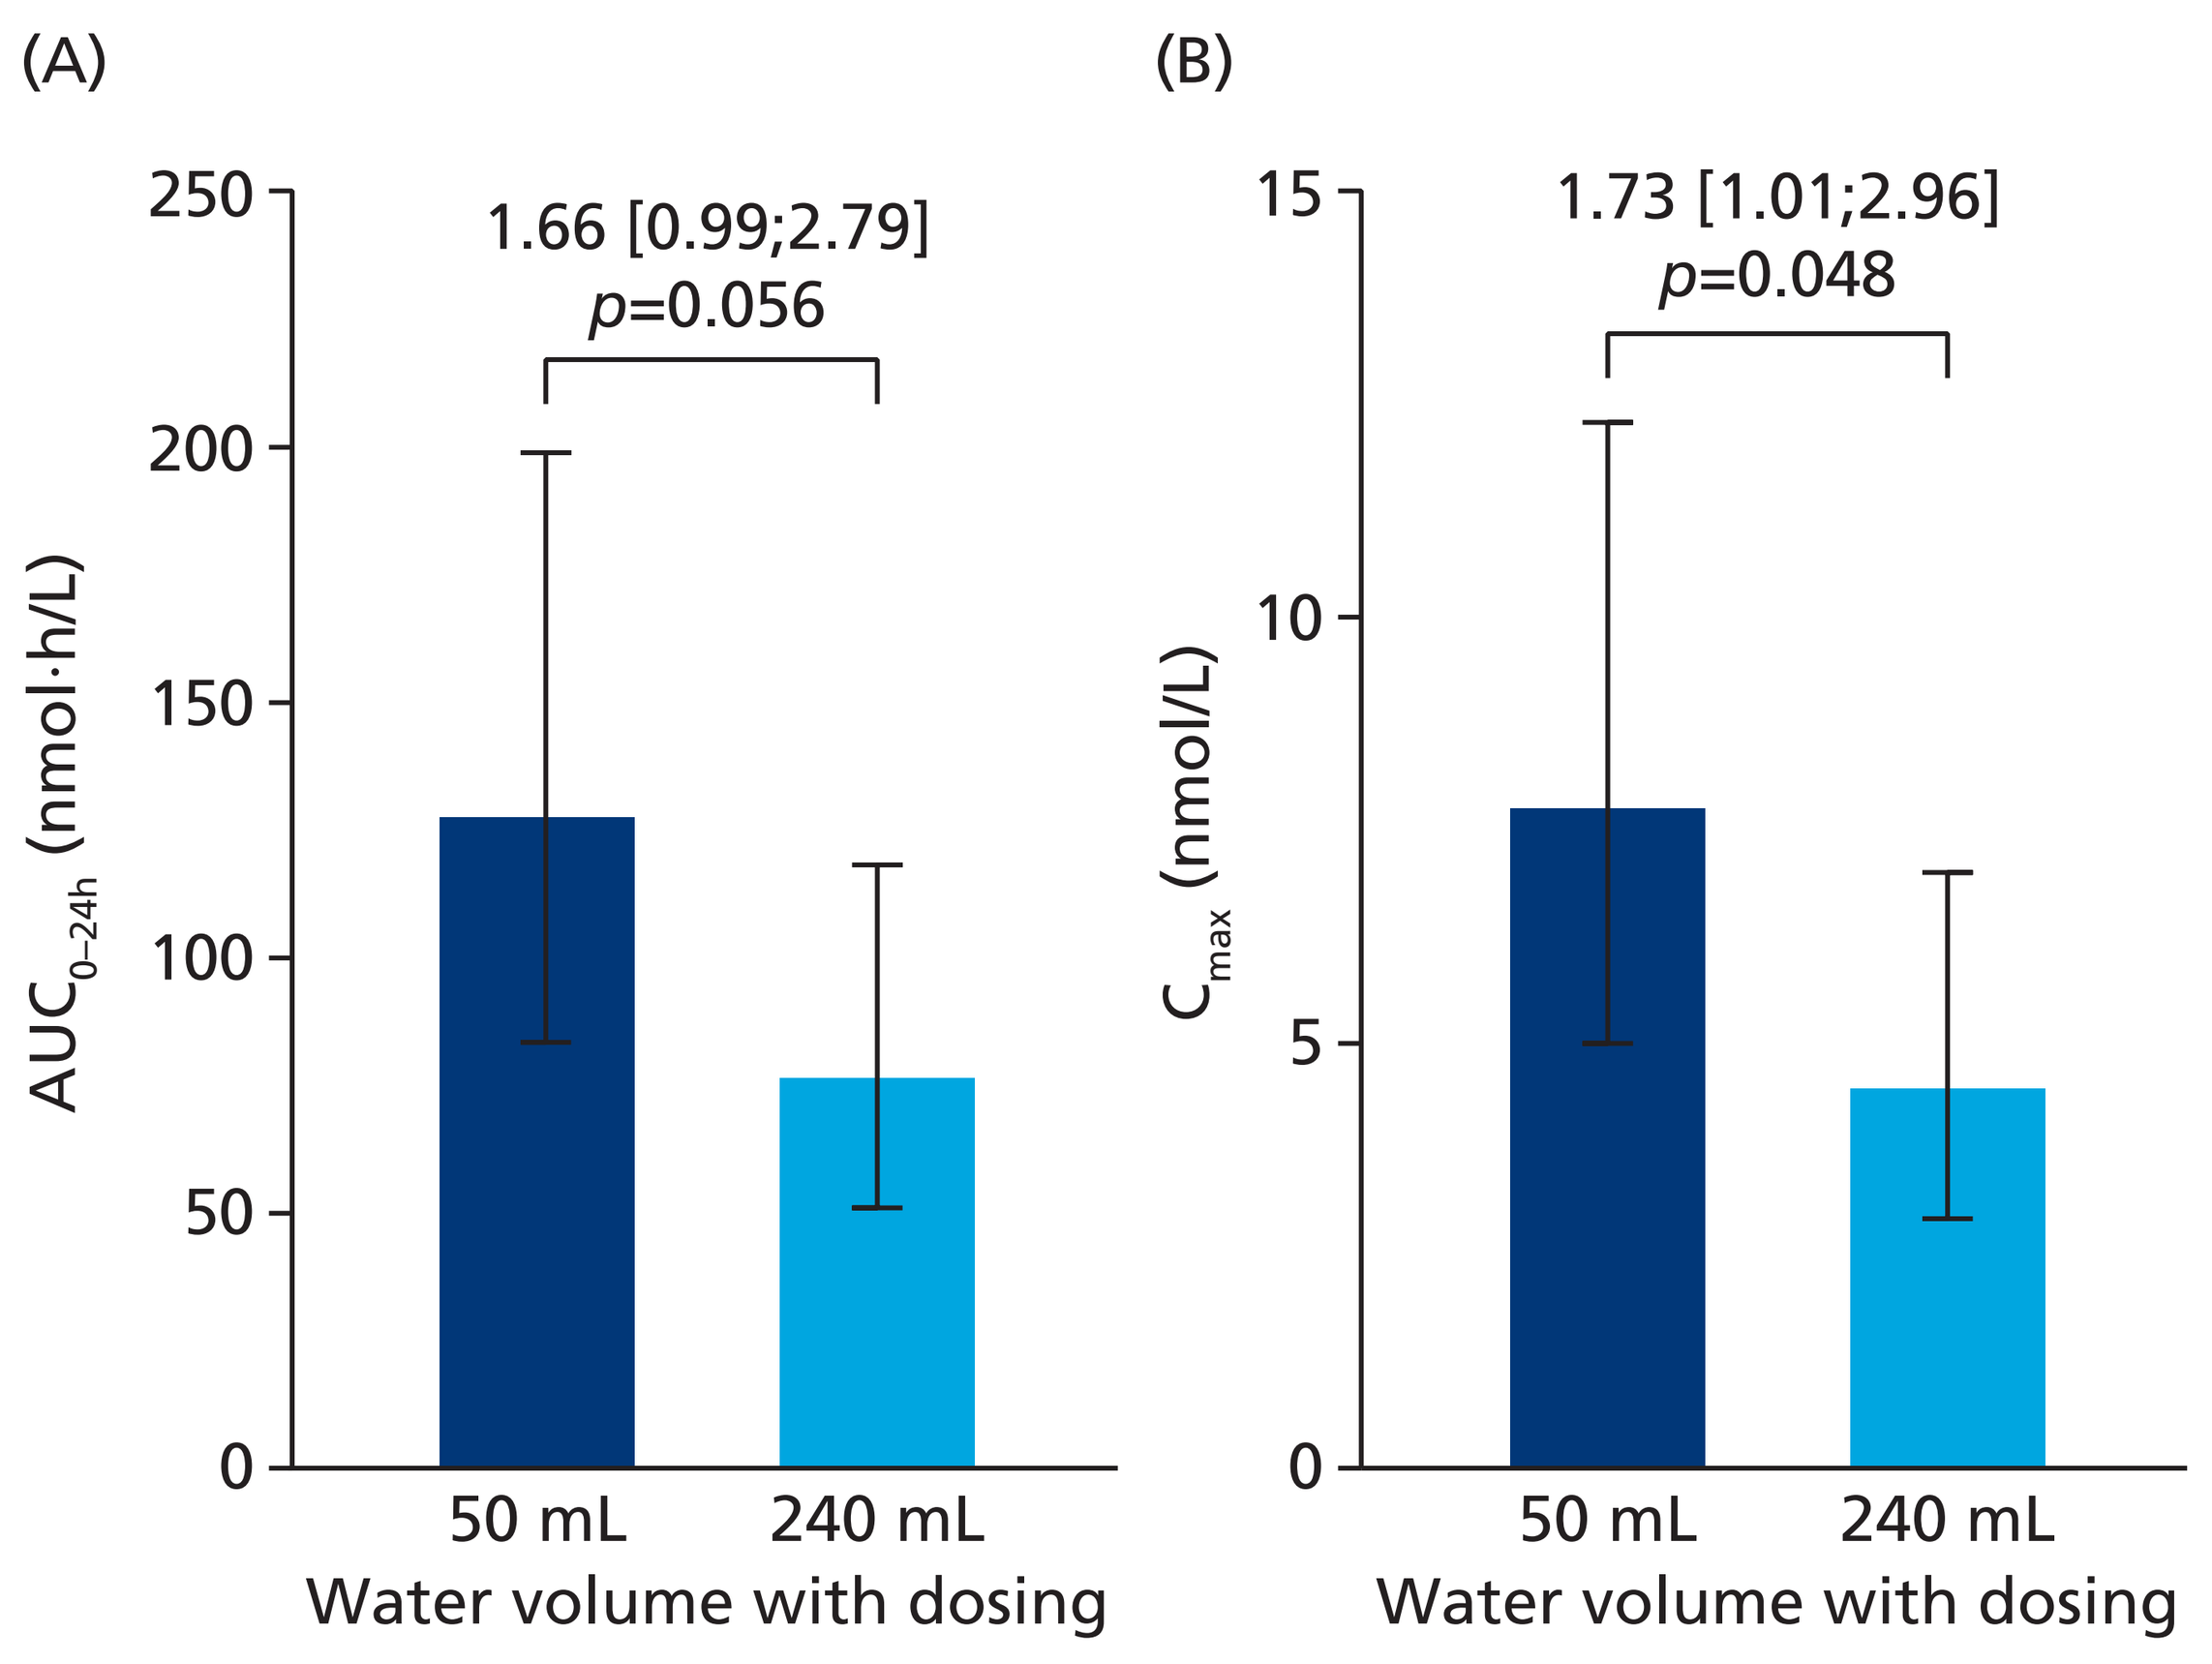

(A)
(B)
AUC0–24h (nmol·h/L)
250
200
150
100
50
0
1.66 [0.99;2.79]
p=0.056
50 mL
240 mL
Water volume with dosing
Cmax (nmol/L)
15
10
5
0
1.73 [1.01;2.96]
p=0.048
50 mL
240 mL
Water volume with dosing

Bars are estimated means and 95% confidence intervals.
Treatment comparisons show estimated treatment ratios [95% confidence interval] and p-value.

Safety

- Overall, the safety profile was as expected for the GLP-1 receptor agonist drug class.
- The most frequently reported adverse events were headache (reported in 17% and 23% of subjects for 50 mL and 240 mL water, respectively), nausea (21% and 12%) and vomiting (17% and 12%).
- The majority of adverse events were mild (47 events), while 7 adverse events were moderate and no adverse events were severe.
- No serious adverse events were reported during the trial.

Conclusions

- Oral semaglutide tablet erosion occurs in the stomach irrespective of water volume.
- A slower rate of tablet erosion in the stomach, as seen when administering the tablet with 50 mL vs. 240 mL of water, and a delayed gastric emptying result in higher semaglutide plasma exposure.

This trial was sponsored by Novo Nordisk A/S and is registered with ClinicalTrials.gov (NCT01619345).
The authors acknowledge the medical writing assistance of CR Pharma Consult.
Presented at the American Diabetes Association, 77th Annual Scientific Sessions, 9–13 June 2017, San Diego, CA, USA.